Neratinib efficacy in a subgroup of patients with *EGFR* exon 18-mutant non-small cell lung cancer and central nervous system involvement: findings from the SUMMIT basket trial

Jonathan W. Goldman,[1] Santiago Viteri Ramirez,[2] Amit Mahipal,[3] Jennifer Marie Suga,[4] Lisa D. Eli,[5] Alshad S. Lalani,[5] Richard Bryce,[6] Feng Xu,[6] Naisargee Shah,[6] Fairooz Kabbinavar,[6] Valentina Boni,[7] Barbara B. Haley[8]

[1]University of California, Los Angeles, Los Angeles, CA, USA; [2]Instituto Oncologico Dr Rosell, Hospital Universitario Dexeus, Grupo Quiron Salud, Barcelona, Spain; [3]Mayo Clinic, Rochester, MN; [4]Kaiser Permanente Cancer Research Center, Vallejo, CA; [5]Puma Biotechnology Inc., San Francisco, CA; [6]Puma Biotechnology Inc, Los Angeles, CA; [7]START Madrid-CIOCC, Hospital Universitario, Madrid Sanchinarro, Madrid, Spain; [8]UT Southwestern Medical Center, Dallas, TX

#9068

Background

- The phase 2 SUMMIT basket trial (NCT01953926) demonstrated efficacy of neratinib in patients with *EGFR* exon 18-mutant non-small cell lung cancer (NSCLC).[1]
- Neratinib also has documented activity in HER2-positive metastatic breast cancer, including patients with central nervous system (CNS) metastases.[2,3]

Objectives

- To report neratinib single-agent efficacy in a cohort of patients with *EGFR* exon 18-mutant NSCLC from SUMMIT, including patients with CNS involvement.

Figure 1. *EGFR* exon 18 mutations represent 5% of all *EGFR* mutations detected in lung cancer

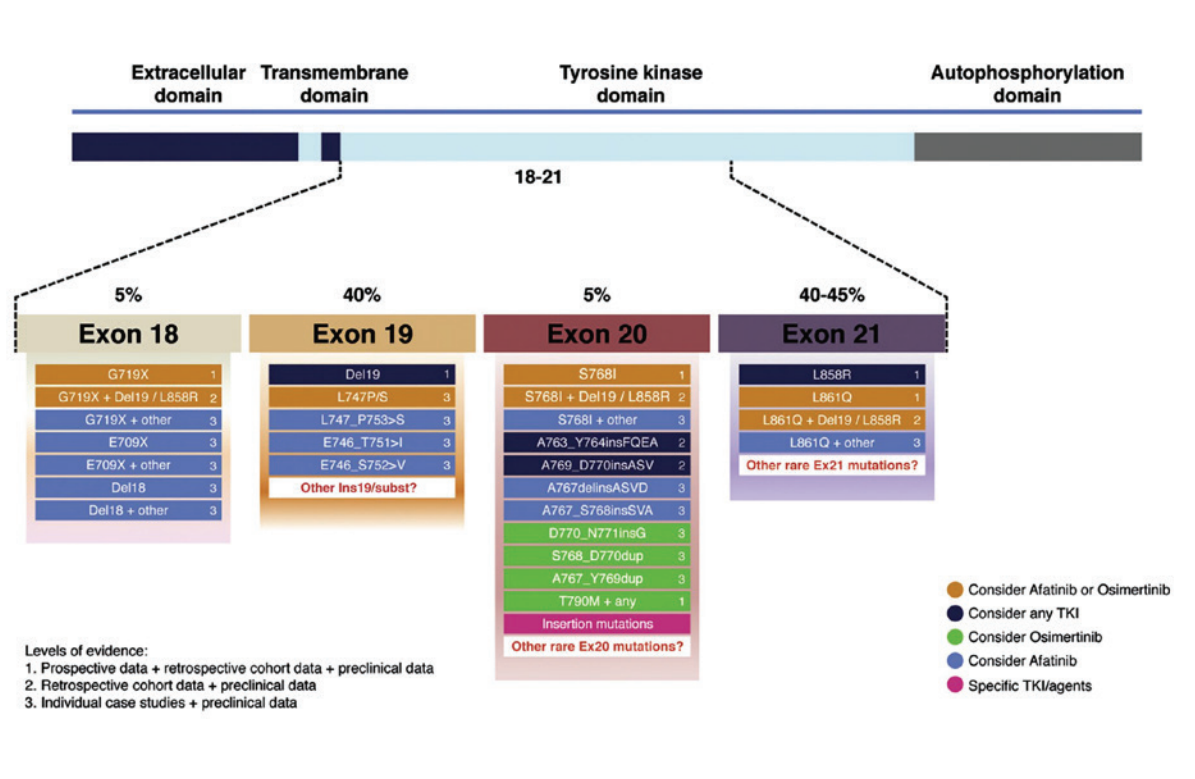

Extracellular domain
Transmembrane domain
Tyrosine kinase domain
Autophosphorylation domain
18-21
5% Exon 18
40% Exon 19
5% Exon 20
40-45% Exon 21
G719X 1
G719X + Del19 / L858R 2
G719X + other 3
E709X 3
E709X + other 3
Del18 3
Del18 + other 3
Del19 1
L747P/S 3
L747_P753>S 3
E746_T751>I 3
E746_S752>V 3
Other Ins19/subst?
S768I 1
S768I + Del19 / L858R 2
S768I + other 3
A763_Y764insFQEA 2
A769_D770insASV 2
A767delinsASVD 3
A767_S768insSVA 3
D770_N771insG 3
S768_D770dup 3
A767_Y769dup 3
T790M + any 1
Insertion mutations
Other rare Ex20 mutations?
L858R 1
L861Q 1
L861Q + Del19 / L858R 2
L861Q + other 3
Other rare Ex21 mutations?
Consider Afatinib or Osimertinib
Consider any TKI
Consider Osimertinib
Consider Afatinib
Specific TKI/agents
Levels of evidence:
1. Prospective data + retrospective cohort data + preclinical data
2. Retrospective cohort data + preclinical data
3. Individual case studies + preclinical data

Reproduced from [4]Passaro A, et al. J Thoracic Oncol 2020; Dec 14:S1556-0864(20)31102-3

Figure 2. *EGFR* exon 18 mutations are highly sensitive to neratinib *in vitro*

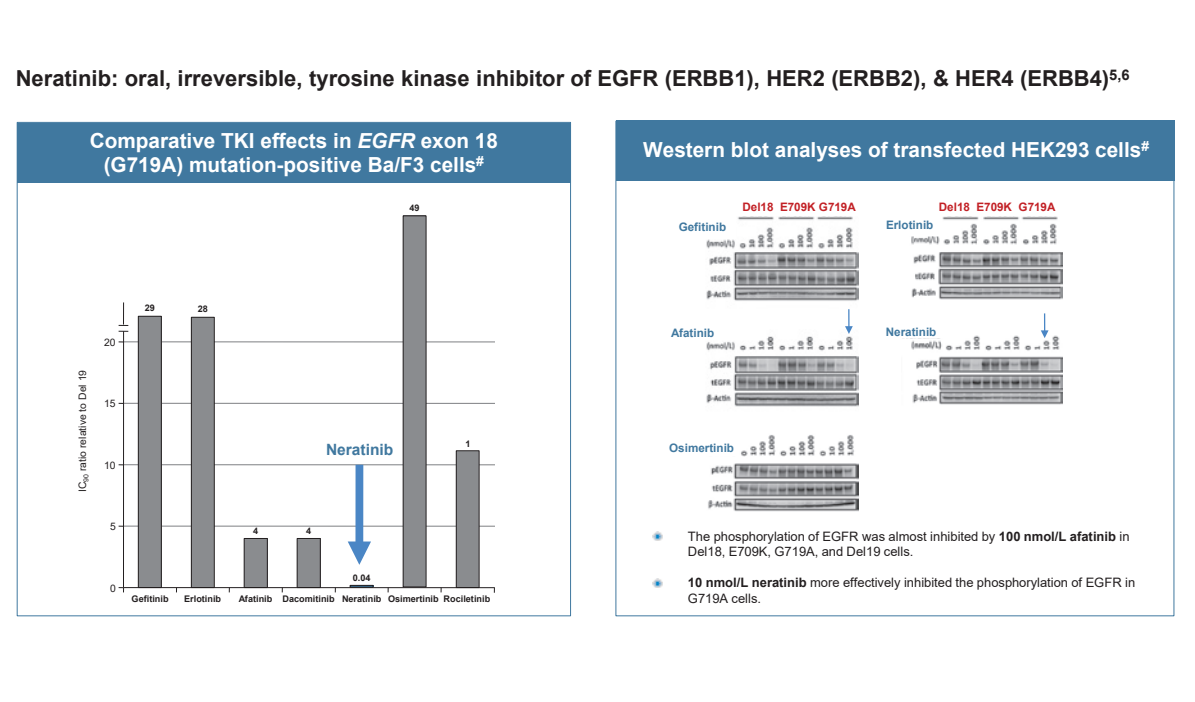

Neratinib: oral, irreversible, tyrosine kinase inhibitor of EGFR (ERBB1), HER2 (ERBB2), & HER4 (ERBB4)5,6
Comparative TKI effects in EGFR exon 18 (G719A) mutation-positive Ba/F3 cells#
Western blot analyses of transfected HEK293 cells#
Neratinib
Gefitinib
Erlotinib
Afatinib
Dacomitinib
Neratinib
Osimertinib
Rociletinib
Del18 E709K G719A
The phosphorylation of EGFR was almost inhibited by 100 nmol/L afatinib in Del18, E709K, G719A, and Del19 cells.
10 nmol/L neratinib more effectively inhibited the phosphorylation of EGFR in G719A cells.

5. Rabindran et al. Cancer Res 2004:64:3958–65; 6. Bose et al. Cancer Discov 2013;3:224–37; #Modified with permission from 7. Kobayashi et al. Clin Cancer Res 2015;21:5305–13

Methods

- The overall SUMMIT study design has been presented previously.[1,9]
- The design of the *EGFR* exon 18-mutant lung cancer cohort is shown in detail in Figure 4.

Figure 3. *EGFR* exon 18 mutations are highly sensitive to neratinib: a POC trial

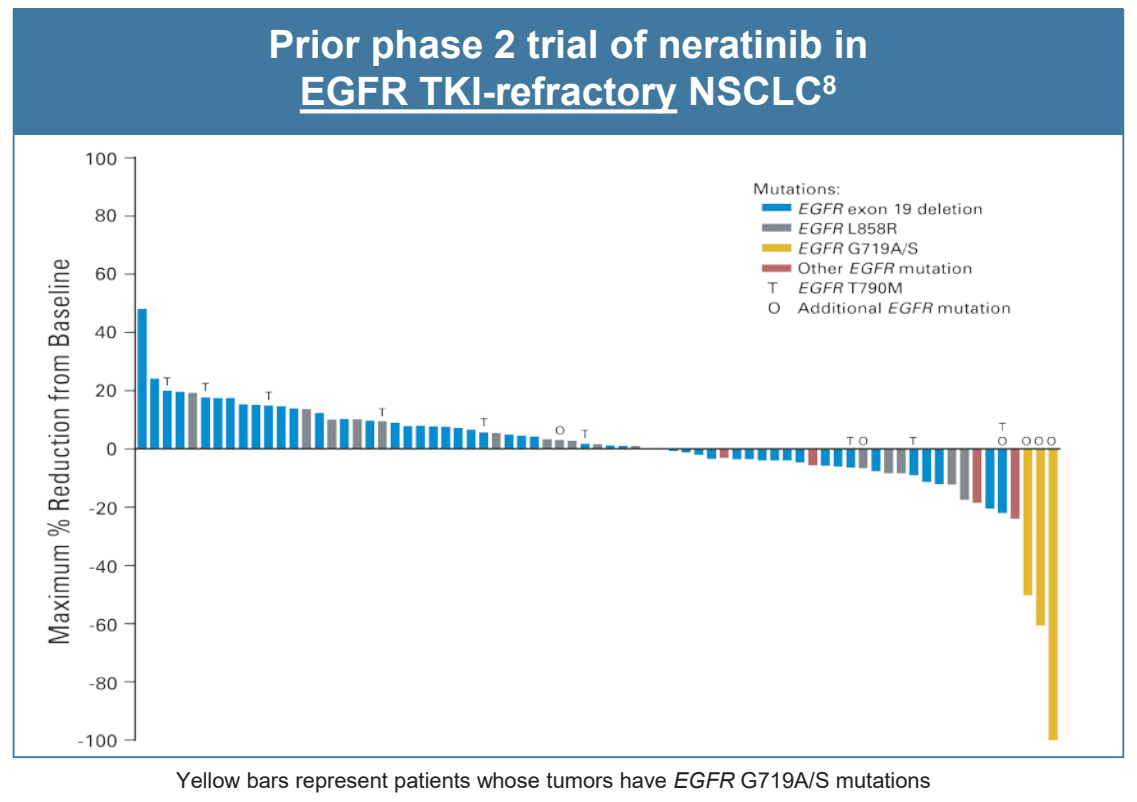

Prior phase 2 trial of neratinib in EGFR TKI-refractory NSCLC8
Maximum % Reduction from Baseline
Mutations:
EGFR exon 19 deletion
EGFR L858R
EGFR G719A/S
Other EGFR mutation
T EGFR T790M
O Additional EGFR mutation

Yellow bars represent patients whose tumors have *EGFR* G719A/S mutations

- 4/167 (2%) patients had *EGFR* exon 18 mutations (*G719X*); 1 patient did not have measurable disease by central review.
- All patients with *G719* mutations (n=4) had clinical benefit: 3 PRs and 1 SD lasting >40 weeks:
 - Median PFS 52.7 weeks (90% CI 25.6–57.0 weeks).

8. Reproduced with permission from Sequist L, et al. J Clin Oncol 2010;28:3076–83.

Figure 4. SUMMIT (A7) study design for *EGFR* exon 18-mutant lung cancer cohort

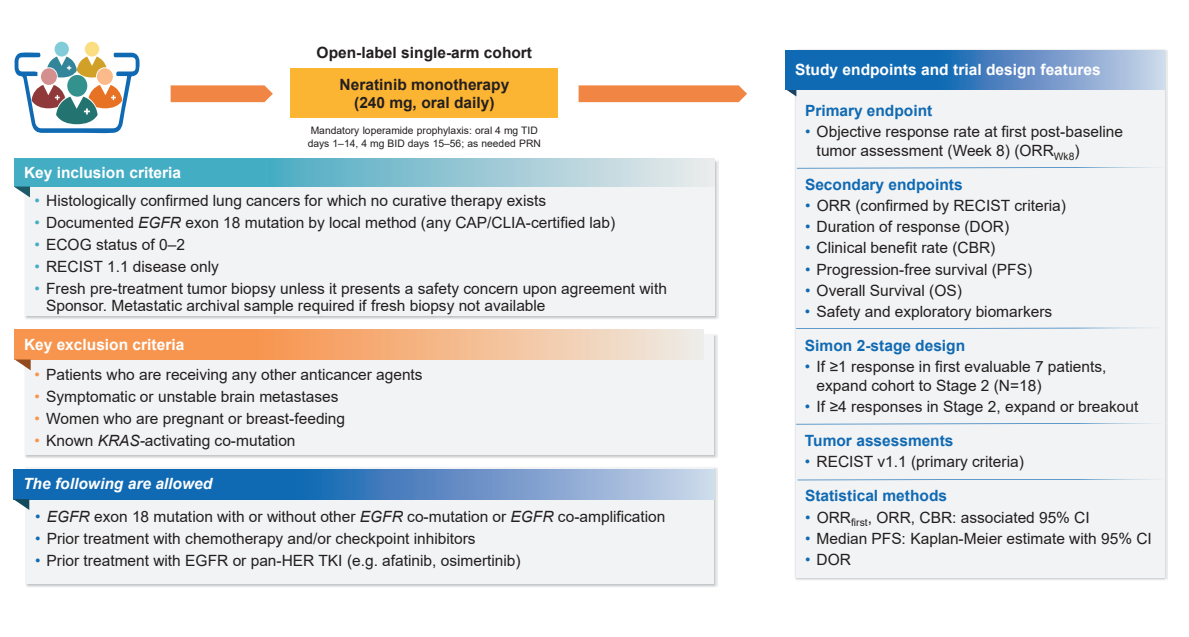

Open-label single-arm cohort
Neratinib monotherapy (240 mg, oral daily)
Mandatory loperamide prophylaxis: oral 4 mg TID days 1–14, 4 mg BID days 15–56; as needed PRN
Key inclusion criteria
Histologically confirmed lung cancers for which no curative therapy exists
Documented EGFR exon 18 mutation by local method (any CAP/CLIA-certified lab)
ECOG status of 0–2
RECIST 1.1 disease only
Fresh pre-treatment tumor biopsy unless it presents a safety concern upon agreement with Sponsor. Metastatic archival sample required if fresh biopsy not available
Key exclusion criteria
Patients who are receiving any other anticancer agents
Symptomatic or unstable brain metastases
Women who are pregnant or breast-feeding
Known KRAS-activating co-mutation
The following are allowed
EGFR exon 18 mutation with or without other EGFR co-mutation or EGFR co-amplification
Prior treatment with chemotherapy and/or checkpoint inhibitors
Prior treatment with EGFR or pan-HER TKI (e.g. afatinib, osimertinib)
Study endpoints and trial design features
Primary endpoint
Objective response rate at first post-baseline tumor assessment (Week 8) (ORRWk8)
Secondary endpoints
ORR (confirmed by RECIST criteria)
Duration of response (DOR)
Clinical benefit rate (CBR)
Progression-free survival (PFS)
Overall Survival (OS)
Safety and exploratory biomarkers
Simon 2-stage design
If ≥1 response in first evaluable 7 patients, expand cohort to Stage 2 (N=18)
If ≥4 responses in Stage 2, expand or breakout
Tumor assessments
RECIST v1.1 (primary criteria)
Statistical methods
ORRfirst, ORR, CBR: associated 95% CI
Median PFS: Kaplan-Meier estimate with 95% CI
DOR

Table 1. Baseline demographics and patient characteristics

Patient characteristics	Safety/Efficacy evaluable patients (n=11)
Median age (range), years	**67** (56–83)
<65 years, n (%)	4 (36)
≥65 years, n (%)	7 (64)
Gender, n (%)	
Female	5 (45)
Male	6 (55)
ECOG performance status, n (%)	
0	5 (45)
1	6 (55)
Race, n (%)	
Black or African American	1 (9)
White	10 (91)
Median number of prior therapies in metastatic/locally advanced setting (range)	2 (1–3)
Prior EGFR tyrosine kinase inhibitor, n (%)	**10** (91)
Prior chemotherapy, n (%)	6 (55)
Prior checkpoint inhibitor, n (%)	3 (27)

Data cutoff date: 21 August 2020

Key efficacy findings

Efficacy in TKI-pretreated patients

- Among 10 TKI-pretreated patients, 60% (n=6) had a PR, and 40% (n=4) had a confirmed PR.
- SD lasting ≥16 weeks was seen in 4 additional patients, giving a CBR of 80%.
- The median DOR with neratinib was 7.5 months.
- The median PFS with neratinib was 9.1 months.
- The median OS with neratinib was 17.9 months.

Efficacy in TKI-pretreated patients with CNS involvement

- 2 patients had PR and 1 patient had SD as best response by RECIST 1.1.
- Individual PFS times were 1.9 (censored), 6.9, and 9.1 months.
- Individual OS times were 2.6 (censored), 17.7 (censored), and 17.9 months.

Table 2. *EGFR* exon 18-mutant lung cancer cohort receiving neratinib monotherapy: Efficacy summary

Parameter	Efficacy-evaluable patients (n=11)	TKI pretreated (n=10)
Objective response (confirmed),[a] n	**4**	**4**
CR	0	0
PR	4	4
Objective response rate, % (95% CI)	**36** (11–69)	**40** (12–74)
Best overall response, n	**6**	**6**
CR	0	0
PR	6	6
Best overall response rate, % (95% CI)	**54** (23–83)	**60** (26–88)
Median DOR,[b] months (95% CI)	**7.5 (4.0–NE) (1.9*, 4.0, 7.5, 9.2*)**	**7.5 (4.0–NE) (1.9*, 4.0, 7.5, 9.2*)**
Clinical benefit,[c] n	**8**	**8**
CR or PR	4	4
SD ≥16 weeks	4	4
Clinical benefit rate, % (95% CI)	**73** (39–94)	**80** (44–97)
Median PFS,[b] months (95% CI)	**6.9** (2.1–NA)	**9.1** (3.7–NA)
PFS in patients with CNS metastases, months	See TKI pretreated	**1.9#, 6.9, 9.1**
Median OS,[b] months (95% CI)	**17.9** (5.8–NE)	**17.9** (5.7–NE)
OS in patients with CNS metastases, months	See TKI pretreated	**2.6#, 17.7#, 17.9**

Data cutoff date: 21 August 2020

[a]Objective response rate (ORR) is defined as either a complete or partial response that is confirmed no less than 4 weeks after the criteria for response are initially met; [b]Kaplan-Meier analysis in safety population; [c]Clinical benefit rate (CBR) is defined as confirmed CR or PR or stable disease (SD) for ≥16 weeks (within ± 7-day visit window); DOR, duration of response; PFS, progression-free survival, *response ongoing

Figure 5. Treatment duration and best response

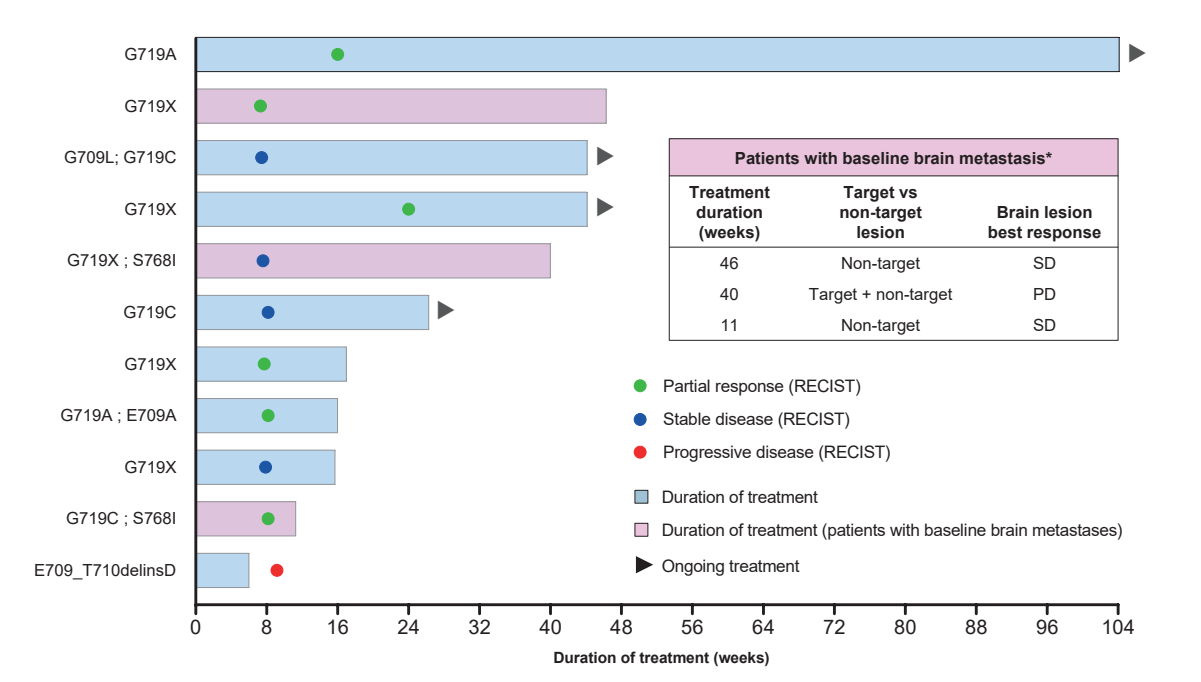

G719A
G719X
G709L; G719C
G719X
G719X ; S768I
G719C
G719X
G719A ; E709A
G719X
G719C ; S768I
E709_T710delinsD
Patients with baseline brain metastasis*
Treatment duration (weeks)
Target vs non-target lesion
Brain lesion best response
46 Non-target SD
40 Target + non-target PD
11 Non-target SD
Partial response (RECIST)
Stable disease (RECIST)
Progressive disease (RECIST)
Duration of treatment
Duration of treatment (patients with baseline brain metastases)
Ongoing treatment
0 8 16 24 32 40 48 56 64 72 80 88 96 104
Duration of treatment (weeks)

Data cutoff date: 21 August 2020

*All patients received palliative radiation to the brain prior to study entry.

Key safety findings

- Neratinib was well tolerated with mandatory loperamide prophylaxis (first 2 cycles).
- Four patients reported grade 1 and one patient reported grade 2 diarrhea (the most common side effect of neratinib).
- There was no evidence of grade 3 diarrhea, interstitial lung disease (ILD), or skin rashes.
- No patients required a dose hold, dose reduction, hospitalization or permanently discontinued neratinib due to diarrhea.

Table 3. *EGFR* exon 18-mutant lung cancer cohort: Most common treatment-emergent adverse events >10%

TEAEs	Safety evaluable patients (n=11)	
	Any grade	Grade ≥3
Diarrhea	5 (45.5)	0
Vomiting	4 (36.4)	0
Constipation	3 (27.3)	0
Nausea	3 (27.3)	0
Decreased appetite	3 (27.3)	1 (9.1)
Dizziness	2 (18.2)	0
Hypertension	2 (18.2)	0
Dry mouth	2 (18.2)	0
Fatigue	2 (18.2)	0

Data cutoff date: 21 August 2020

Case study

- Female, 61 years, former smoker.
- December 2016: stage IV lung adenocarcinoma with lung, lymph nodes, bone and brain metastasis and *EGFR* (G719X) mutation.
- December 2016: SBRT on brain; 1st-line erlotinib achieving SD as best response and clinical benefit.
- November 2018: asymptomatic brain/lung progression; 2nd-line neratinib (duration of treatment 46 weeks).
- **January 2019: PR (60% reduction in tumor burden by RECIST 1.1) and stable brain mets on neratinib.**
- September 2019: lung PD; 3rd-line osimertinib.

Baseline Nov 2018 **Jan 2019**

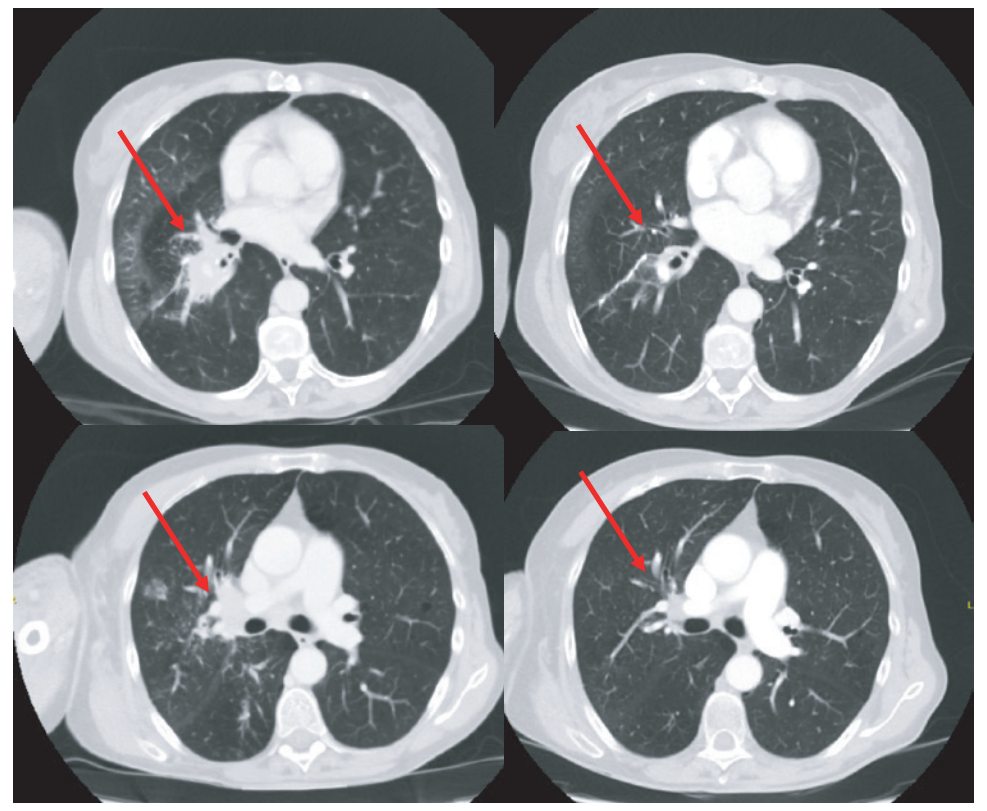

Conclusions

- Activity of single-agent neratinib was observed in prior TKI-exposed patients with *EGFR* exon 18-mutant NSCLC.
- Despite the small sample size of only 3 patients with baseline CNS metastases, the current findings suggest a potential role for neratinib as a systemic treatment option for patients with NSCLC and difficult-to-treat uncommon mutations with CNS involvement:
 - These data are consistent with the CNS activity of neratinib in HER2-overexpressing breast cancer.
- Neratinib single-agent treatment was well tolerated in *EGFR* exon 18-mutant NSCLC patients, with no evidence of grade ≥3 diarrhea.
- In addition, no ILD or skin rash was noted.
- The SUMMIT trial continues to enroll patients with *EGFR* exon 18-mutant NSCLC, with or without prior TKI.

References

1. Boni V, et al. WCLC 2020 (OA04).
2. Saura C, et al. SABCS 2020 (PD13-09).
3. Saura C, et al. J Clin Oncol 2020;38:3138–49.
4. Passaro A, et al. J Thoracic Oncol 2020; Dec 14:S1556-0864(20)31102-3.
5. Rabindran SK, et al. Cancer Res 2004:64:3958–65.
6. Bose R, et al. Cancer Discov 2013;3:224–37.
7. Kobayashi Y, et al. Clin Cancer Res 2015;21:5305–13.
8. Sequist L, et al. J Clin Oncol 2010;28:3076–83.
9. Hyman DM, et al. Nature 2018;554:189–94.

Acknowledgements

- The authors would like to thank all patients and their families for participating in the SUMMIT trial.
- SUMMIT was sponsored by Puma Biotechnology Inc.
- Puma Biotechnology Inc. funded medical writing/editing assistance for this poster, which was provided by Miller Medical Communications Ltd.

Conflict of interest disclosures: Jonathan W. Goldman

Financial interests

- *Consulting or advisory role:* Amgen, AstraZeneca, Bristol-Myers Squibb, Genentech/Roche, Lilly.
- *Speakers' bureau:* Merck.
- *Travel, accommodation, expenses:* AstraZeneca.
- *Research funding:* Abbvie, Advaxis, Array BioPharma, AstraZeneca/MedImmune, Bristol-Myers Squibb, Corvus Pharmaceuticals, Genentech/Roche, Lilly, Puma Biotechnology, Spectrum Pharmaceuticals.

Copyright 2021 Puma Biotechnology

Presented at the 2021 ASCO Annual Meeting, June 4–8, 2021 (virtual)

This presentation is the intellectual property of the author/presenter. Contact them at Email: jwgoldman@mednet.ucla.edu for permission to reprint and/or distribute