Effects of adding budesonide or colestipol to loperamide prophylaxis on neratinib-associated diarrhea in patients with HER2+ early-stage breast cancer: the CONTROL trial

Sara Hurvitz,[1] Arlene Chan,[2] Nicholas Iannotti,[3] Emad Ibrahim,[4] Jo Chien,[5] Nancy Chan,[6] Andrew Kellum,[7] Vincent Hansen,[8] Gavin Marx,[9] S. DiSean Kendall,[10] Mary Wilkinson,[11] Aurelio Castrellon,[12] Rolando Ruiz,[13] Pearl Fang,[13] Daniel Hunt,[13] Susan Moran,[13] Elizabeth Olek,[13] Carlos H. Barcenas,[14] Hope S. Rugo[5]

[1]UCLA Hematology / Oncology Clinical Research Unit, Los Angeles, CA, USA; [2]Breast Cancer Research Centre-WA & Curtin University, Perth, WA, Australia; [3]Hematology Oncology Associates of the Treasure Coast, Port Saint Lucie, FL, USA; [4]Center for Surgical and Specialty Care, Redlands, CA, USA; [5]UCSF Comprehensive Cancer Center, San Francisco, CA, USA; [6]Rutgers Cancer Institute of New Jersey, New Brunswick, NJ, USA; [7]North Mississippi Medical Center Hematology and Oncology Services, Tupelo, MS, USA; [8]Northern Utah Associates, Ogden, UT, USA; [9]Sydney Adventist Hospital, Wahroonga, NSW, Australia; [10]Utah Cancer Specialists Research, Salt Lake City, UT, USA; [11]Inova Medical Group, Fairfax, VA, USA; [12]Memorial Healthcare System, Hollywood, FL, USA; [13]Puma Biotechnology Inc., Los Angeles, CA, USA; [14]MD Anderson Cancer Center, Houston, TX, USA

#P3-14-01

Background

- Neratinib, an irreversible tyrosine kinase inhibitor of HER1, HER2 and HER4,[1] is used for the extended adjuvant treatment of early-stage HER2-positive (HER2+) breast cancer after trastuzumab-based adjuvant therapy.
 - The international, randomized, placebo-controlled phase III ExteNET trial showed that a 12-month course of neratinib after trastuzumab based adjuvant therapy significantly improved invasive disease free survival compared with placebo after 2 years (hazard ratio 0.67; 95% CI 0.50–0.91; p=0.0091)[2] and 5 years (hazard ratio 0.73; 95% CI 0.57–0.92; p=0.008).[3]
- Diarrhea is the main toxicity of neratinib and is common in the absence of proactive management.[2]
 - As most diarrhea events with neratinib occur early in the course of treatment,[2] a structured (intensive) prophylactic regimen of loperamide given for 1–2 cycles is recommended with neratinib to better manage this toxicity.[4]
 - Preclinical studies suggest that neratinib-associated diarrhea may be multifactorial with possible inflammatory, secretory and bile acid malabsorption etiologies (Puma data on file).
 - In a rat model of pan-HER tyrosine kinase inhibitor-induced diarrhea, a reduction in diarrhea was achieved with an anti-inflammatory agent or a bile acid resin.[5]
- CONTROL is an international, open-label, sequential-cohort, phase II study investigating the effects of loperamide prophylaxis alone or with add-on budesonide, a locally acting corticosteroid used for inflammatory gastrointestinal conditions, or colestipol, a bile acid sequestrant, on neratinib-associated diarrhea.
- We report updated safety findings and the first health-related quality-of-life data (HRQoL) from the CONTROL study.

Methods

- CONTROL (PUMA-NER-6201) is an international, sequential-cohort, open-label, phase II study.
- The study is registered (Clinicaltrials.gov identifier NCT02400476).

Patient population

- Adults ≥18 years of age.
- Histologically confirmed stage 1–3c breast cancer.
- Documented HER2 overexpression or amplification determined locally.
- Completed trastuzumab-based adjuvant therapy, or experienced side effects resulting in early discontinuation, with last trastuzumab dose given >2 weeks and ≤1 year prior to enrollment.

Study treatment

- Details of treatment schedules by cohort are presented in Figure 1.
 - Cycle length = 4 weeks.
- All eligible patients received:
 - Oral neratinib 240 mg/day for 1 year.
 - Oral loperamide prophylaxis for 1 or 2 cycles.
 - Loperamide (≤16 mg/day) as needed after completion of loperamide prophylaxis.
- Patients in the budesonide and colestipol cohorts also received add-on oral budesonide or colestipol for 1 cycle, respectively.
- In addition, any treatment-emergent diarrhea was managed with:
 - Neratinib dose modifications, i.e. dose holds or dose reductions according to a protocol defined schedule.
 - Dietetic measures and additional pharmacological treatments depending on grade (i.e. diphenoxylate plus atropine, octreotide, IV fluids, antibiotics).
- In patients unable to tolerate loperamide, loperamide + budesonide, or loperamide + colestipol due to symptomatic constipation, the loperamide dose could be held until after the first bowel movement and then resumed at a reduced dose.

Figure 1. Treatment schedules by cohort

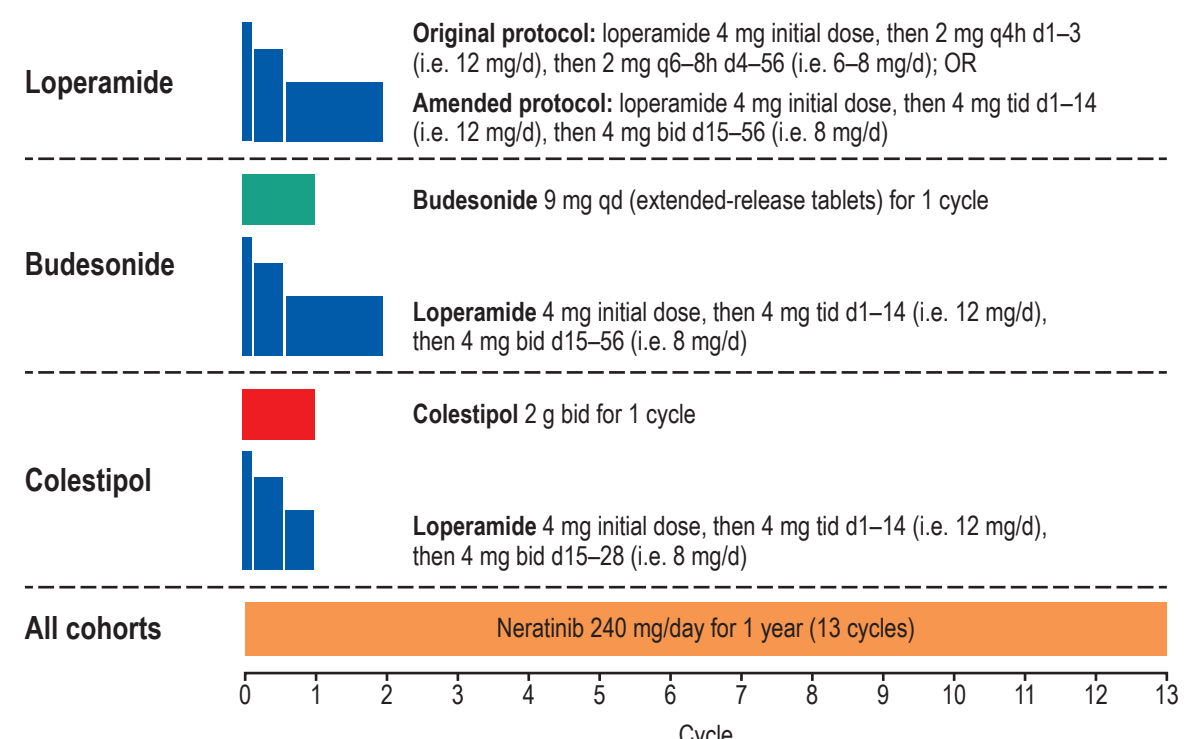
Loperamide
Original protocol: loperamide 4 mg initial dose, then 2 mg q4h d1–3 (i.e. 12 mg/d), then 2 mg q6–8h d4–56 (i.e. 6–8 mg/d); OR
Amended protocol: loperamide 4 mg initial dose, then 4 mg tid d1–14 (i.e. 12 mg/d), then 4 mg bid d15–56 (i.e. 8 mg/d)
Budesonide
Budesonide 9 mg qd (extended-release tablets) for 1 cycle
Loperamide 4 mg initial dose, then 4 mg tid d1–14 (i.e. 12 mg/d), then 4 mg bid d15–56 (i.e. 8 mg/d)
Colestipol
Colestipol 2 g bid for 1 cycle
Loperamide 4 mg initial dose, then 4 mg tid d1–14 (i.e. 12 mg/d), then 4 mg bid d15–28 (i.e. 8 mg/d)
All cohorts
Neratinib 240 mg/day for 1 year (13 cycles)
0 1 2 3 4 5 6 7 8 9 10 11 12 13
Cycle

One cycle = 28 days
Abbreviations: bid, twice daily; tid, 3-times daily; qd = once daily
CONTROL new cohort administering colestipol and loperamide PRN; newly planned cohort will administer neratinib in a dose escalation over the first cycle, to 240 mg qd for 1 year.

Endpoints

- *Primary:* incidence of grade ≥3 diarrhea.
- *Secondary:* frequency distribution of maximum-grade diarrhea; incidence and severity of diarrhea by loperamide exposure; serious adverse events; adverse events of interest.
- *Exploratory:* patient-reported HRQoL; exploratory biomarkers.

Assessments

- Clinic visits were scheduled on day 1 of cycles 1, 2, 3, 4, 7 and 10, and treatment end.
- Patients were contacted by phone at 1 day, 2 days and 3 days after the first dose of neratinib and all patients were required to use a diary to record intake of neratinib, loperamide and any other medications throughout the study.
- Patients were followed for 28 days after the last dose of neratinib.
- Adverse events were graded according to NCI-CTCAE (version 4.0).

HRQoL

- HRQoL was assessed using Functional Assessment of Cancer Therapy for Breast Cancer (FACT-B), version 4.0, and EuroQoL 5-Dimensions 5-level (EQ-5D-5L); only data for FACT-B are presented here.
 - FACT-B is a 37-item questionnaire with 5 subscales assessing physical, social, emotional, and functional well-being, as well as a breast cancer-specific subscale.
- HRQoL questionnaires were completed electronically by patients on day 1 of cycles 1, 2, 3, 4, 7 and 10, and end of treatment.
- HRQoL assessments were introduced in November 2015 (protocol amendment 2).

Statistical methods

- All safety analyses were descriptive and were performed in the safety population, defined as all patients who received ≥1 dose of neratinib.
- HRQoL analyses were also descriptive and were performed in the QoL analysis population, defined as all patients in the safety population with a baseline and at least 1 post-baseline QoL assessment. Mean (± standard error) observed scores over time were presented as plots by cohort.
 - Changes in HRQoL scores from baseline or between groups were considered to be clinically meaningful if greater than the lowest estimate for an "important difference" (ID) reported in the literature.
- The ExteNET trial, which included an analogous patient population but no protocol-mandated antidiarrheal prophylaxis,[2] was used as an historical control.
- Cut-off date for the current analysis: November 2017.

Results

- Between 27 February 2015 and 3 November 2017, a total of 321 patients were enrolled from 41 sites: loperamide cohort (n=137); budesonide cohort (n=64); colestipol cohort (n=120).
- Baseline characteristics are presented in **Table 1**.

Table 1. Baseline characteristics

	CONTROL			ExteNET
Variable	Loperamide cohort (n=137)	Budesonide cohort (n=64)	Colestipol cohort (n=120)	(n=1420)
Female, %	100	100	98	100
Median age (range), years	53 (30–86)	49 (29–78)	53 (26–78)	52 (25–83)
Tumor stage at diagnosis,[a] %				
I	28.5	25.0	16.7	9.8
IIA, B	54.7	46.9	46.7	42.0
IIIA, B, C	14.6	23.4	26.7	31.2
IV	0.7	0	0.8	0
Hormone receptor status, %				
Positive (ER+ and/or PR+)	75.2	71.9	72.5	57.5
Negative (ER– and PR–)	24.8	28.1	26.7	42.5
Missing	0	0	<1	0
Prior (neo)adjuvant therapy, %				
Trastuzumab	99.3	96.9	99.2	100
Taxanes	95.6	96.9	98.3	77.3
Anthracycline	26.3	28.1	24.2	90.1
Pertuzumab	40.1	60.9	62.5	–
Median (range) duration of prior trastuzumab, months	11.5 (2.4–18.2)	10.9 (9.8–11.6)	11.0 (10.0–11.8)	11.5 (0.7–56.9)
Median (range) time since last trastuzumab dose, months	3.9 (0.1–12.1)	4.3 (0.5–17.1)	2.7 (0.0–18.6)	4.4 (0.2–30.9)

ER, estrogen receptor; PR, progesterone receptor.

Neratinib exposure

- All patients in the loperamide cohort have now either completed planned neratinib treatment or prematurely discontinued treatment.
 - As of November 2017, 73% of patients in the budesonide cohort and 21% of patients in the colestipol cohort have completed treatment.
- Median duration of neratinib treatment in the loperamide, budesonide and colestipol cohorts is 11.5, 11.9 and 3.7 months, respectively.

Figure 2. Worst grade of treatment-emergent diarrhea (Safety population)

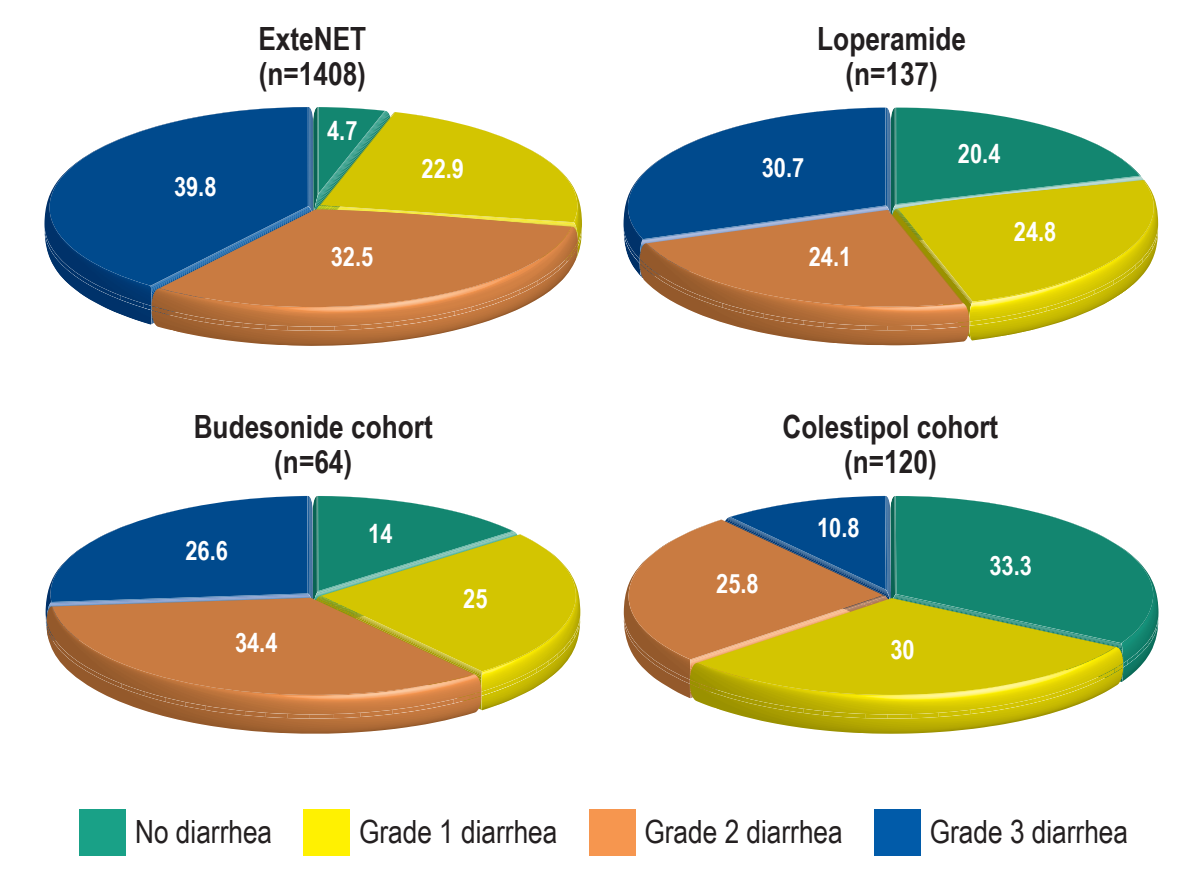
ExteNET (n=1408)
4.7
22.9
32.5
39.8
Loperamide (n=137)
20.4
24.8
24.1
30.7
Budesonide cohort (n=64)
14
25
34.4
26.6
Colestipol cohort (n=120)
33.3
30
25.8
10.8
No diarrhea
Grade 1 diarrhea
Grade 2 diarrhea
Grade 3 diarrhea

Treatment-emergent diarrhea

- Incidence of grade ≥3 diarrhea, the primary study endpoint (**Figure 2**):
 - Loperamide: 30.7% (95% CI 23.1–39.1)
 - Loperamide with budesonide: 26.6% (95% CI 16.3–39.1)
 - Loperamide with colestipol: 10.8% (95% CI 5.9–17.8)
 - ExteNET historical control: 39.9% (95% CI 37.3–42.5).
- There were reductions in the median cumulative duration of diarrhea and in the median number of diarrhea episodes per patient with loperamide prophylaxis given with or without budesonide or colestipol *vs* ExteNET (**Table 2**).
- The proportion of patients requiring neratinib holds and dose reductions was reduced with loperamide prophylaxis given with or without budesonide or colestipol *vs* ExteNET.
- Neratinib discontinuations due to diarrhea in the first cycle (month 1) were lower in the budesonide and colestipol cohorts compared to the loperamide cohort.

Table 2. Characteristics of treatment-emergent diarrhea (Safety population)

Study	CONTROL			ExteNET[3]
Variable	Loperamide (n=137)	Loperamide + budesonide (n=64)	Loperamide + colestipol (n=120)	Loperamide prn (n=1408)
Median cumulative duration, days				
Any grade	14.0	24.0	16.0	59.0
Grade ≥2	5.0	6.0	3.5	10.0
Grade ≥3[a]	3.0	2.0	3.0	5.0[a]
Median diarrhea episodes/patient				
Any grade	2	9	2.5	8
Grade ≥2	2	3	1	3
Grade ≥3[a]	1	1	1	2
Action taken, %				
Dose hold	15.3	18.8	9.2	33.9
Dose reduction	7.3	3.1	4.2	26.4
Discontinuation	20.4	10.9	1.7	16.8
Hospitalization	1.5	0	0	1.4

[a]One grade 4 event reported in ExteNET.

- The occurrence and severity of diarrhea in the CONTROL study over the course of neratinib treatment was markedly reduced from that observed in the ExteNET study:
 - ExteNET showed a profile for diarrhea that was chronic and characterized by grades 2 and 3 diarrhea with the greatest incidence in cycle 1 (month 1); and still observed in months 2–12.
 - In the CONTROL study cohorts, the incidence of grade 2–3 diarrhea was reduced during cycle 1 (month 1) and was also reduced in months 2–12.
 - There appears to be some adaptation to the effects of neratinib, as higher-grade diarrhea occurs early during the course of treatment and is less common as treatment continues.

Other adverse events

- Aside from diarrhea, the overall tolerability profile of neratinib with loperamide prophylaxis given with or without budesonide or colestipol was similar to that reported in the ExteNET trial, apart from an increase in constipation.
 - Rates of grade 1/2 constipation in the loperamide, budesonide and colestipol cohorts were 42.3%/14.6%, 62.5%/12.5%, and 53.3%/9.2%, respectively.
 - No grade 3 or higher constipation has been observed to date.
- The most frequently reported grade 3/4 events are shown in **Table 3**.
- Reported grade 4 events (serious adverse events) were sepsis and urinary tract infection (both unrelated events in the same patient); there were no fatal adverse events reported.

HRQoL

- Mean change from baseline in FACT-B total scores by visit for the loperamide CONTROL cohort compared with the ExteNET study are shown in **Figure 3**.
- In the loperamide CONTROL cohort there was some impact on HRQoL, which then resolved towards baseline as in the ExteNET study.
- All changes in FACT-B total scores from baseline reported in the CONTROL study were less than the important difference range (7–8 points).[6]

Table 3. Most common grade 3 or 4 treatment-emergent adverse events (≥1% total incidence in CONTROL)

	CONTROL			ExteNET
Grade 3/4 events, %	Loperamide cohort (n=137)	Budesonide cohort (n=64)	Colestipol cohort (n=120)	Neratinib arm (n=1408)
Diarrhea	30.7	26.6	10.8	39.9
Fatigue	3.6	7.8	1.7	1.6
Vomiting	1.5	3.1	1.7	3.3
Abdominal pain	1.5	1.6	0.8	1.7
Dehydration	1.5	1.6	0.8	0.9

Figure 3. Mean change from baseline in FACT-B total scores by CONTROL cohort and for ExteNET

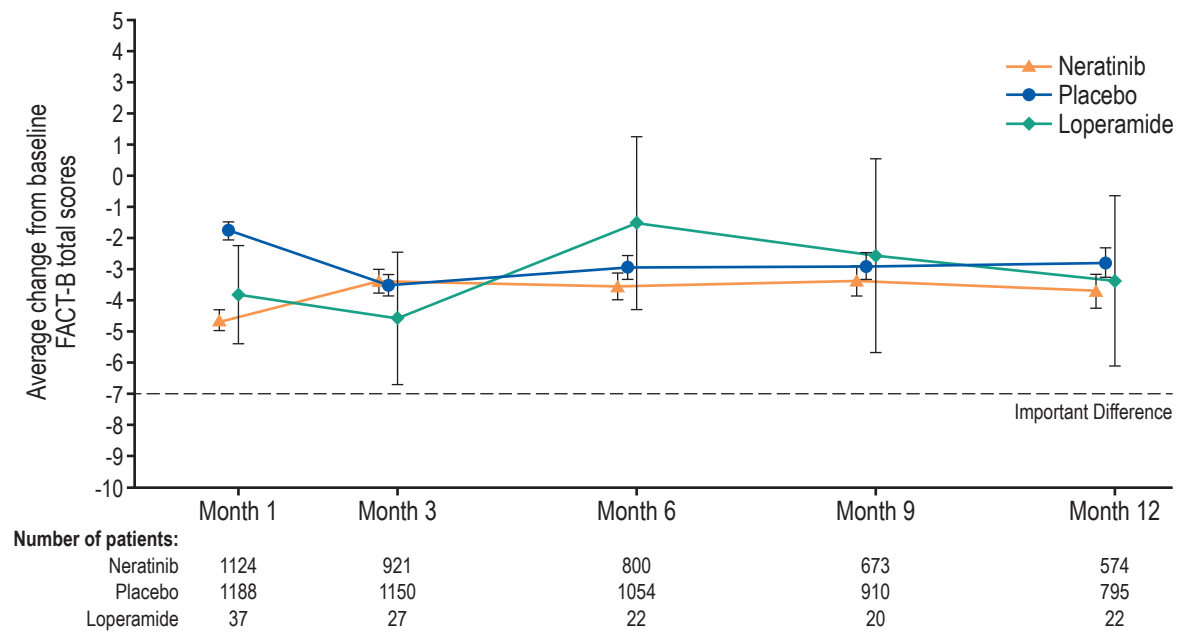
Average change from baseline FACT-B total scores
5
4
3
2
1
0
-1
-2
-3
-4
-5
-6
-7
-8
-9
-10
Neratinib
Placebo
Loperamide
Important Difference
Month 1
Month 3
Month 6
Month 9
Month 12
Number of patients:
Neratinib 1124 921 800 673 574
Placebo 1188 1150 1054 910 795
Loperamide 37 27 22 20 22

Conclusions

- The addition of colestipol to loperamide prophylaxis resulted in the greatest reduction in diarrhea incidence and severity compared to that observed in the ExteNET trial, and may further diminish the duration of diarrhea.
 - Colestipol may also improve tolerability as shown by the decreased rate of other adverse events (including fatigue, headache, abdominal pain) and fewer neratinib dose holds, dose reductions, and discontinuations.
 - Further follow-up is necessary in the ongoing budesonide and colestipol cohorts.
- A structured loperamide prophylactic regimen for one or two cycles with or without the addition of either budesonide or colestipol for a single cycle, reduces the incidence, severity, and duration of neratinib-associated diarrhea compared to that observed in the ExteNET trial.
- Any HRQoL impairment was short-lived and did not reach predefined clinically meaningful thresholds in the loperamide cohort. However, the small sample size and lack of within-study comparator arm limits the conclusions that can be drawn from these data.
- Effective diarrhea prophylaxis may help to improve the tolerability of neratinib, enhance long-term adherence to treatment, and ensure that the efficacy benefits of neratinib are realized.
- The final analysis of the CONTROL study will be performed when all patients have completed the planned 12 months of neratinib therapy.

References

1. Rabindran SK. Cancer Lett 2005;227:9–23.
2. Chan A, et al. Lancet Oncol 2016;17:367–77.
3. Martin M, et al. Lancet Oncol 2017, in press.
4. Ustaris R, et al. Am J Hematol/Oncol 2015;11:13–22.
5. Puma Biotechnology Inc, data on file.
6. Eton DT, et al. J Clin Epidemiol 2004;57:898–910.

Acknowledgements

CONTROL was sponsored by Puma Biotechnology Inc. Puma Biotechnology Inc. also funded the provision of editorial support provided by Miller Medical Communications.

Copyright 2017 Puma Biotechnology

Presented at the 40th Annual San Antonio Breast Cancer Symposium (SABCS), Dec 5–9, 2017, San Antonio, TX